Detection of Silent UTI: A Case Study

H. HUA[1], D. SINISCALCHI[1], D. POWER[1], J. GRAVES[2]
[1]OCTAVE, MENLO PARK, CA, USA; [2]UNIVERSITY OF CALIFORNIA SAN DIEGO, SAN DIEGO, CA, USA

MOC04

Introduction

- Persons with MS (PwMS) have a higher risk of developing urinary tract infections (UTIs) due to bladder dysfunction, use of catheters, increased disability, and use of immunomodulatory and immunosuppressive treatments. UTIs are associated with higher hospitalization rates and mortality rates in this population [1].
- Prevention and management of UTIs, including early diagnosis and treatment, patient education, and timely access to specialists care are needed to reduce UTI related hospitalizations [2].
- Symptomatic UTIs can usually be detected. Silent or asymptomatic UTIs don't present with typical urinary symptoms. They are often masked by neurological worsening and if untreated can progress to serious complications and hospitalizations [3].
- Octave's Clinical Insights (CI) Program utilizes a collaborative care approach to actively and remotely monitor PwMS. Using mobile technology and interaction with a Nurse Care Partner, who is MS certified, the program offers a solution for tracking symptoms, side effects, and clinical changes over time.

Objective

To describe a real-world case study using the Octave CI Program for identification and management of silent UTIs in PwMS and prevention of UTI-associated complications.

Case Description

53 YO Woman | Diagnosed in 2011 | Ambulatory with existing bilateral foot numbness/ tingling | 1–2 UTIs per year | Dimethyl fumarate since 2012

She reported worsening of her left foot numbness/tingling, and severe right low back pain 6 months post total knee replacement (TKR). She attributed these symptoms to a recent episode of extensive walking and a swollen knee from her TKR. She was prescribed muscle relaxants after a consultation with an outside provider with minimal improvement. Frequent live chats and follow-up video calls with a Nurse Care Partner raised the possibility of a silent UTI. The participant was adamant that a silent UTI was not the issue and declined additional work-up. With persistent support and education, she eventually agreed to lab work and clinic follow-up. This led to diagnosis, treatment and resolution of the silent UTI, as well as the back pain within 3 weeks. The participant acknowledged that without the consistent interaction within Octave's CI program, she would have ignored the suggestion of a silent UTI and may have ended up in the emergency room with serious complications.

Timelines

Timeline 1: Events and interactions between patient and Octave Nurse Care Partner (NCP)

- **08/06/2021** - Enrolled in Octave's Clinical Insights MOVING-MS study
- **09/20/2021** - Left Total Knee Replacement (procedure symptoms attributed to)
- **03/10/2022** - First report of increased symptoms (new back pain and increased foot numbness/tingling)
- **03/14/2022** – 3/24/2022 - Ongoing discussion between patient and NCP regarding symptoms; recommended UTI screening
- **03/25/2022** - Patient visit with NCP; reports UA was positive for UTI (labs showed moderate bacteria, 2+ nitrites and protein)
- **03/28/2022** - Patient had visit with PCP; starts a 14-Day course of antibiotics
- **03/29/2022** - Patient reports symptoms improving
- **05/04/2022** - Visit with NCP; patient reports symptoms completely resolved

Timeline 2: Longitudinal symptom severity and duration captured by the Octave CI Program

Key Symptoms Over Time 1 New And 1 Ongoing

Back Pain

Mild to None March 2022 – May 2022

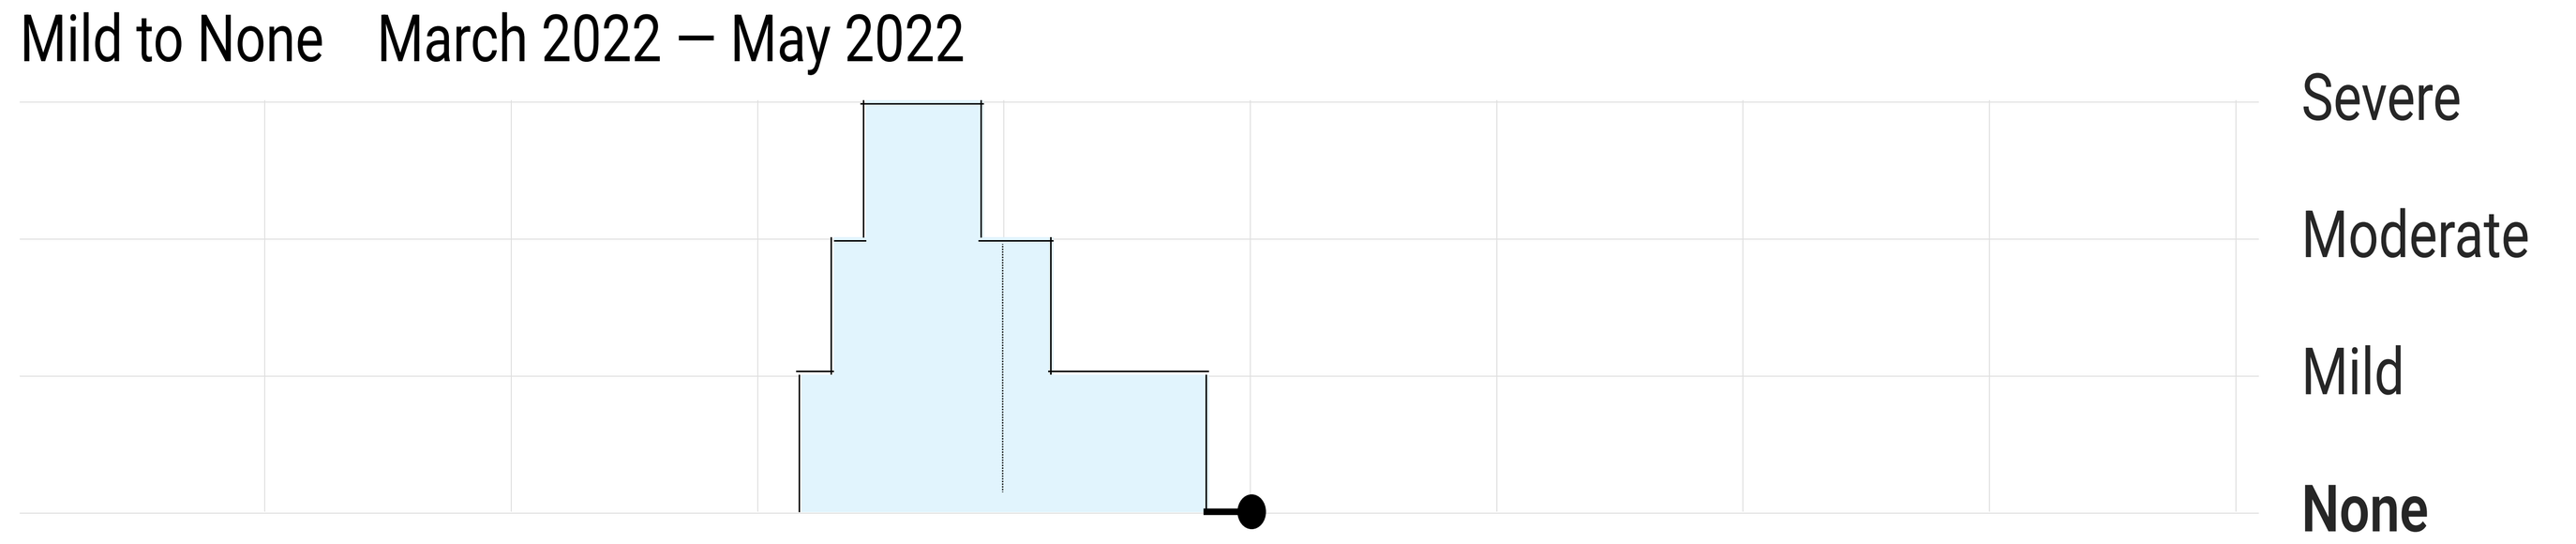
Severe
Moderate
Mild
None

New

- 3/10/22 sudden acute low back pain 3 days post trip to Las Vegas
- Resolved within 2 weeks of antibiotic completion

Left foot/arch numbness/tingling/tightness

Moderate to Mild Oct 2021 – Today

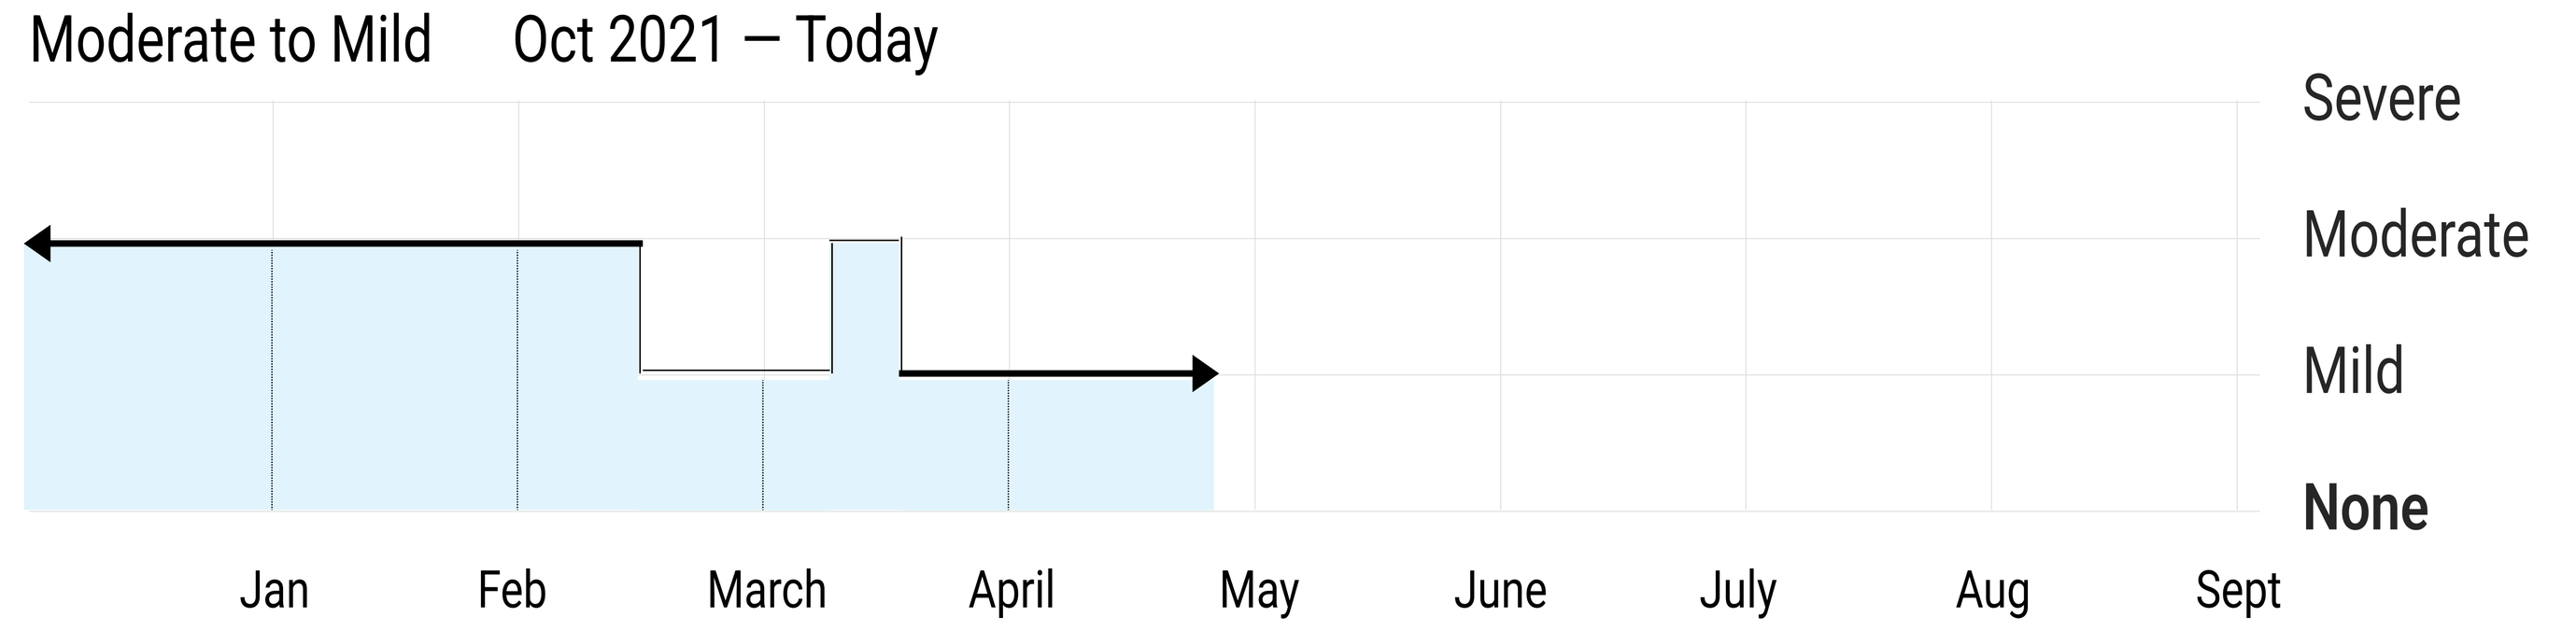
Severe
Moderate
Mild
None
Jan
Feb
March
April
May
June
July
Aug
Sept

Changed

- Ongoing mild MS symptom
- Worsened post total knee replacement
- Exacerbated by severe silent UTI
- Returned to baseline following antibiotic completion

Results

Screener Outcomes

Confirmed Relapse
- No events

Pseudo Relapse
- None

Confirmed Infection (2)
- Symptomatic UTI 8/27/21
- Silent UTI 3/28/22
- Both resolved following antibiotics

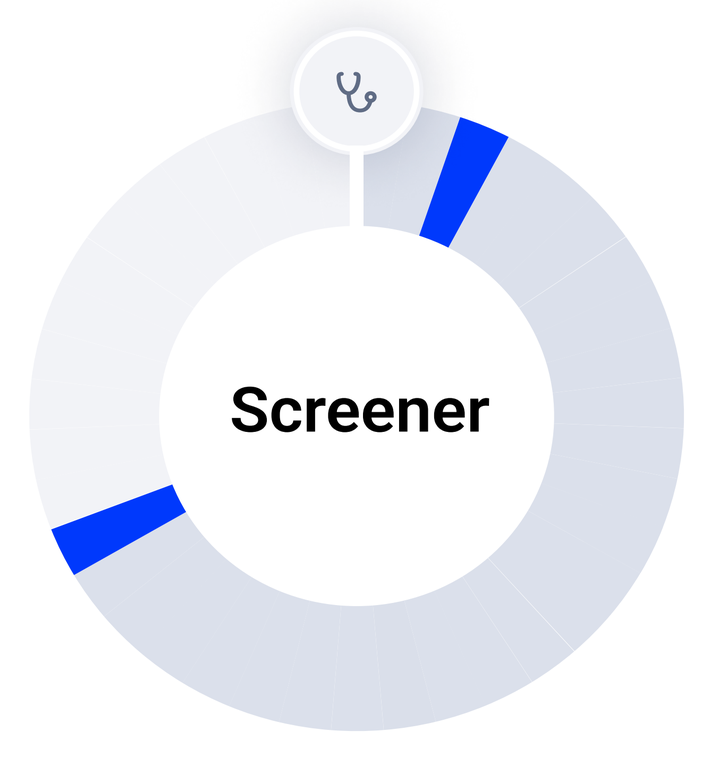
Screener
Aug 2021 - May 2022

Figure 1: Screener graph depicts the number of relapse, pseudo relapse, and infections confirmed from Octave CI Program by the Nurse Care Partner

"Thank you so much for listening and giving me guidance on all of my health needs. Seeing me through my UTIs, COVID, and total knee replacement. I am glad I was part of the study and hope people will use it and find it helpful." *- KK*

Conclusions

- Octave's CI Program combines the use of mobile monitoring and direct nursing interaction and may allow for early intervention in the detection of silent UTIs.
- Access to routine specialized care is necessary for comprehensive management of PwMS. There may be missed opportunities when non-specialized providers are unable to connect symptoms back to MS, especially in the case of silent UTIs that do not present with the typical urinary symptoms.
- Several barriers exist in healthcare utilization that contribute to the reluctance of PwMS to seek medical attention. They often choose not to reach out to busy clinics and assign low priority to their symptoms with the hope that they will resolve over time. An advocate, such as the Nurse Care Partner can be an integral component of comprehensive care.
- Future studies will determine healthcare utilization and patient/physician satisfaction in a larger sample enrolled in the Octave MOVING-MS Trial.

References

[1] Washington, L.G., Demore, C.C., Mazaro, L.P., Souza, M., Parolin, L. Melo, L., Werka Jr. C., Gonçalves, M.(2020) Urinary tract infection in patients with multiple sclerosis: An overview. https://pubmed.ncbi.nlm.nih.gov/32890816/
[2] Li, V., Barker, N., Curtis, C., Porter, B., Panicker, J. N., Chataway, J., & Pakzad, M. (2020). The prevention and management of hospital admissions for urinary tract infection in patients with multiple sclerosis. Multiple sclerosis and related disorders, 45, 102432. https://doi.org/10.1016/j.msard.2020.102432
[3] Jordan, K., Hua, H., & Edgeworth, M. Urinary tract infection in people with multiple slerosis: A real-world administrative claims study. The 8th annual meeting of the American Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS), February 23-25, 2023, San Diego, CA. Urinary Tract Infections in People with Multiple Sclerosis: A Real-World Administrative Claims Study

DISCLOSURES: This study was funded by Octave Bioscience. HH, DS, and DP are employees of Octave Bioscience. JG is an employee of The University of California, San Diego. She has received research support from Octave.

Presented at Consortium of Multiple Sclerosis Centers (CMSC), May 31 - June 3, 2023 in Aurora, Colorado